ResearchGate

See discussions, stats, and author profiles for this publication at: https://www.researchgate.net/publication/308598402

Comparison of four in silico models to predict cardiac action potential effects and proarrhythmia liability

Article · September 2016

DOI: 10.1016/j.vascn.2016.02.050

CITATIONS
0

READS
22

3 authors:

Annie Delaunois
Union Chimique Belge (UCB)
58 PUBLICATIONS **447** CITATIONS

SEE PROFILE

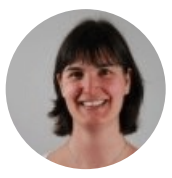

Aurore Colomar
Université de Mons
15 PUBLICATIONS **235** CITATIONS

SEE PROFILE

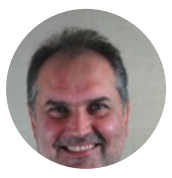

Jean-Pierre Valentin
UCB Biopharma
65 PUBLICATIONS **108** CITATIONS

SEE PROFILE

Some of the authors of this publication are also working on these related projects:

Project cardiac safety detection and human stem-cell derived cardiomyocytes View project

All content following this page was uploaded by Annie Delaunois on 04 January 2017.

The user has requested enhancement of the downloaded file.

Comparison of four *in silico* models to predict cardiac action potential effects and pro-arrhythmia liability

Annie Delaunois [1], Aurore Colomar [1], Jean-Pierre Valentin [1]

[1] Investigative Toxicology, Non-Clinical Development, UCB Biopharma SPRL, Belgium

Poster n°49

Introduction: CiPA goal and core components

- The **Comprehensive in vitro Proarrhythmia Assay (CiPA)** is a novel cardiac safety screening paradigm intended to replace the current regulatory strategy based on the S7B and E14 guidance. It utilizes high throughput methods and provides a more complete and accurate assessment of pro-arrhythmia potential in man, going beyond QT prolongation or TdP (Sager et al. (2014) Am. Heart J.,167(3):292-300)
- The consequence for pharmaceutical companies would be to obtain a waiver for TQT study, provided that:
 - ✓ Non-clinical data from S7A core studies and from CiPA assays have identified a low pro-arrhythmia risk
 - ✓ Robust QT data are collected in Phase I clinical trials
- Three primary components allowing electrophysiological understanding of pro-arrhythmia:

In vitro **functional effects on multiple human cardiac ion currents (patch clamp experiments)**

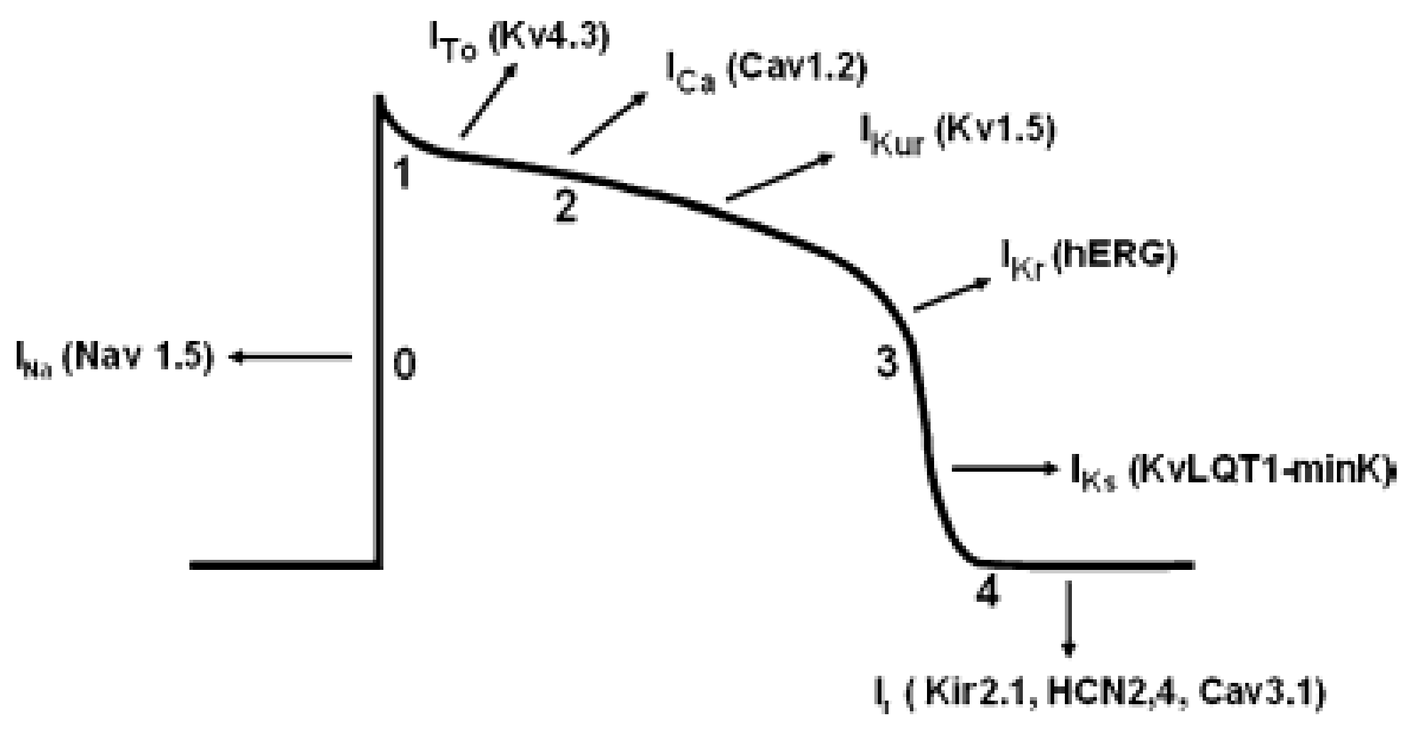
I_{To} (Kv4.3)
I_{Ca} (Cav1.2)
I_{Kur} (Kv1.5)
I_{Kr} (hERG)
I_{Na} (Nav 1.5)
I_{Ks} (KvLQT1-minK)
I_f (Kir2.1, HCN2,4, Cav3.1)
0
1
2
3
4

Cardiac ion currents
(*from Brown, Eur. Biophys. J., 2009*)

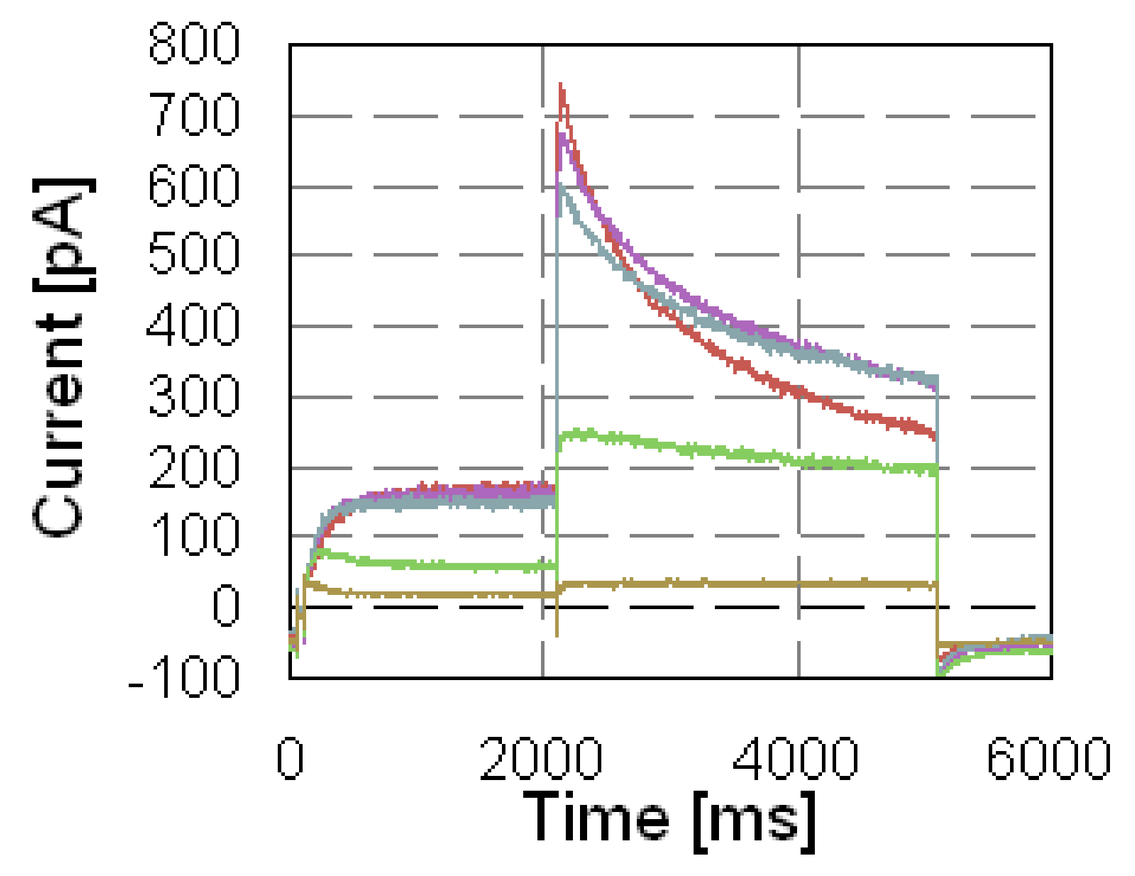
Current [pA]
Time [ms]

hERG current in a patch clamp (*UCB data*)

In silico **cellular simulation of electrophysiology effects (action potential)**

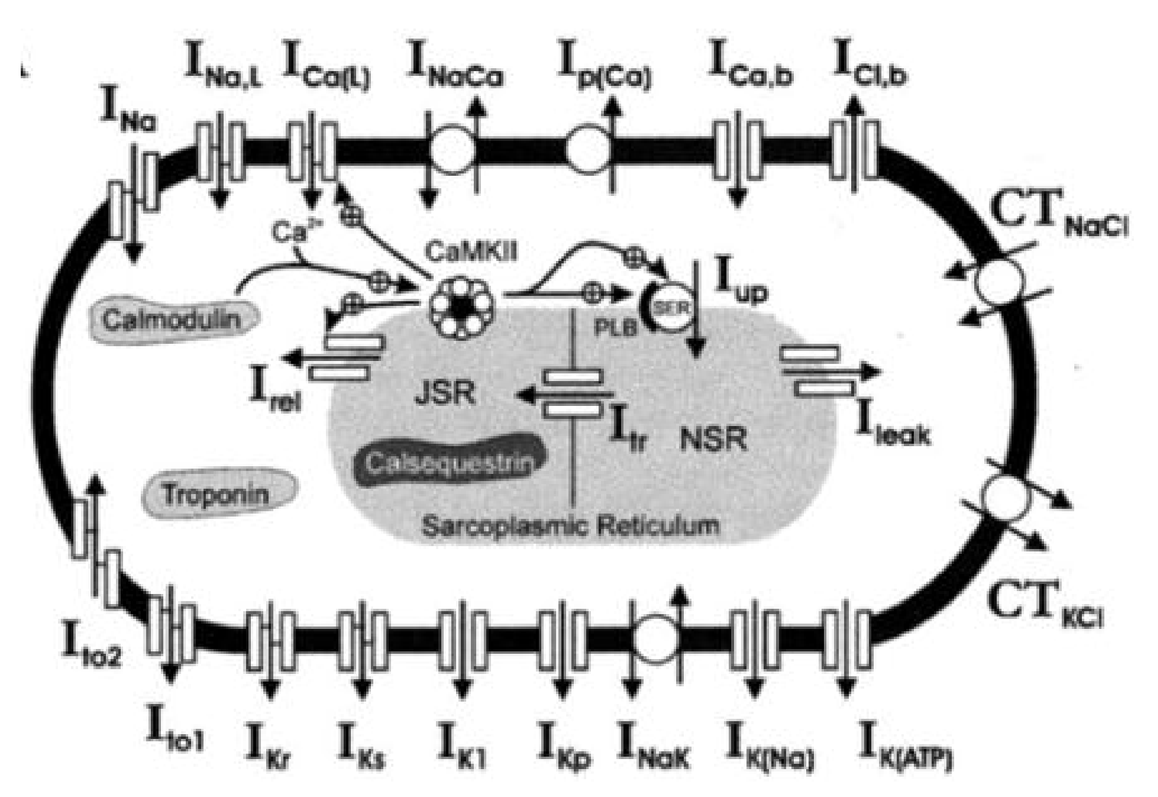
Calmodulin
CaMKII
JSR
NSR
Calsequestrin
Sarcoplasmic Reticulum
Troponin
CT_{NaCl}
CT_{KCl}

(*from Hund & Rudy, Circ., 2004*)

In vitro **effects on human stem cells-derived ventricular myocytes**

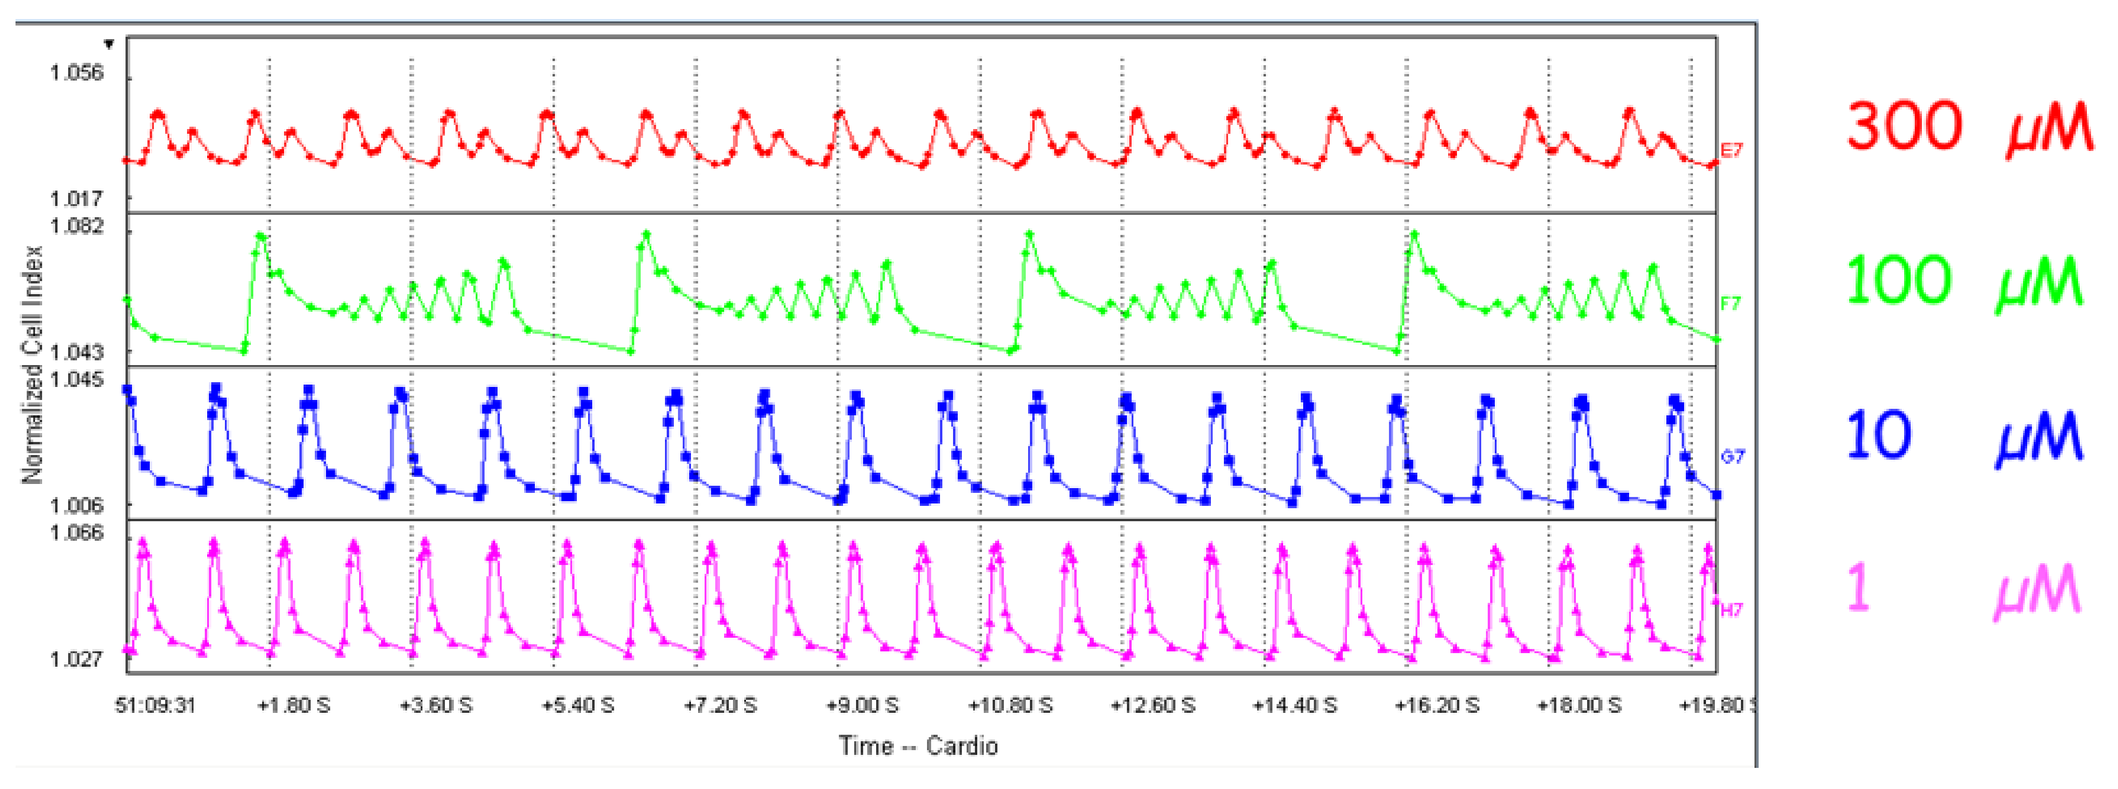
300 µM
100 µM
10 µM
1 µM
Normalized Cell Index
Time -- Cardio

Contraction profiles after flecainide treatment (*UCB data*)

In silico models: comparative work

- Various *in silico* tools have been developed over the last few years and are now proposed by academic partners or private providers
- We first compared four of these models based on the different criteria described here below:

Criteria	In silico model A	In silico model B	In silico model C	In silico model D
Number of channels/states integrated	Up to 3	Up to 6	Up to 15	Up to 6
Number of parameters generated	1 ($APD_{30-40-50-70-90}$) with more in development	1 (APD_{90}) with possibility for more (RMP, Tr)	8 ($APD_{40-60-90}$, APA, RMP, Tr_{40-60}, EAD, TDR, RUD, Vmax)	6 (APD_{50-90}, QRS interval, QT/QTc interval, T wave peak/end, JT interval, EMW)
Number of tested species	3 (dog, rabbit, man)	1 (dog only)	4 (dog, rabbit, guinea pig, man)	1 (human only)
Cell type	Ventricular myocytes	Ventricular myocytes	Ventricular and atrial myocytes, Purkinje fibers	Ventricular myocytes (epi-, midmyo-, endocardial)
Choice of experimental conditions	No	Yes (cycle length, extracellular concentration in Na^+, K^+, Ca^{2+})	Yes (cycle length, extracellular concentration in Na^+, K^+, Ca^{2+})	Yes (gender, age, cardiomyocyte area/volume, capacitance, string length, simulation period, diffusion coefficient, extracellular concentration in Na^+, K^+, Ca^{2+})
Direct access by the customer (on-line platform)	Yes	No	No	Yes
Type of data used for model validation	Literature data	Lliterature and proprietary data	Literature data	Literature and proprietary data
Publication(s) on the model	Yes (poster at SPS 2014 by Fernandez et al., JPTM 70 (3):321)	Yes (Davies et al., Am J Physiol Heart Circ Physiol 302:H1466, 2012)	Yes (Christophe, Pharm Rep 65:1281, 2013)	No (related publications by Polak et al., 2014-2015)

APA: action potential amplitude; APD: action potential duration; EAD: early-after depolarization; EMW: electromechanical window; RMP: resting membrane potential; RUD: reverse use-dependence; TDR: transmural dispersion of repolarization; Tr: triangulation; Vmax: maximal depolarization velocity

- We also compared simulation results generated by the 4 models on 3 UCB compounds for which a full data set of ion currents, Purkinje fibers, and clinical data were available:

	In silico model A ([1] dog for X-Y, [2] rabbit for Z)	In silico model B ([3] dog for all)	In silico model C ([4] dog for X-Y, [5] rabbit for Z)	In silico model D ([6] human)	Real world data
Compound X	**from 0.01 to 0.3µM**: ↑ **APD** then, from 1 to **30µM: ↓ APD**	from 0.3 to **1µM: ↑ APD**; then, From 3 to **30µM: ↓ APD**	**from 0.1 to 30µM**: ↑ **APD** also ↓ **APA, ↓ Vmax**, ↓ Tr	**From 0.1µM: ↓ APD** and ↑ EMW From 1µM: **shortened** QTc	Dog PF: no effects at 0.3µM; ↑ APD at 3µM with RUD; ↓ APD, APA, Vmax at 30µM Man: prolonged QTc, TdP
Compound Y	**from 0.01 to 1µM: ↑ APD** then ↓ **APD** from 3 to 100µM	**no change at 0.1-1µM**; ↓ **APD from 10** to 100µM	From 0.1 to 100µM: ↑ **APD** From 1 to 100µM:↓ APA, ↓ **Vmax** From 10 to 100µM: ↓ **Tr**	Not tested	Dog PF: no effects at 0.1-1µM; ↓ APD and Tr from 10µM; ↓ Vmax and abnormal AP at 100µM Man: atrial pauses, AV blocks II, syncope at 0.015µM
Compound Z	**from** 0.1 to **10µM: ↑ APD** then ↓ **APD from 30** to 100µM	**no change at 0.1-1µM**; ↑ **APD from 10** to 100µM	From 1 to 100µM: ↑ **APD**, ↑ or ↓ Tr From 10 to 100µM:↓ APA, ↓**Vmax**	Not tested	Rabbit PF: ↑ APD with EAD from 10µM; ↓ Vmax at 30µM Guinea pig telemetry: prolonged QTc and PR at 2µM

Red: discrepancy with real world data; green: concordance with real world data.
Algorithms used: [1] Benson et al. (2008) Prog Biophys Mol Biol 96:187-208; [2] Shannon et al. (2004) Biophys J 87:3351; [3] adapted from Benson et al. (2008) and Hund & Rudy (2004) Circ 110:3168; [4] Aslanidi et al. (2009) Biophys J 97:20; [5] Aslanidi et al. (2010) Biophys J 98:2420; [6] adapted from Ten Tusscher et al. (2004) Am J Phys Heart Circ Phys 286: H1573-89 and O'Hara et al. (2011) PLoS Comput Biol 7(5):e1002061, Negroni and Lascano (2008) J Mol Cel Cardiology 45(2):300-12

MODEL A – Compound Y

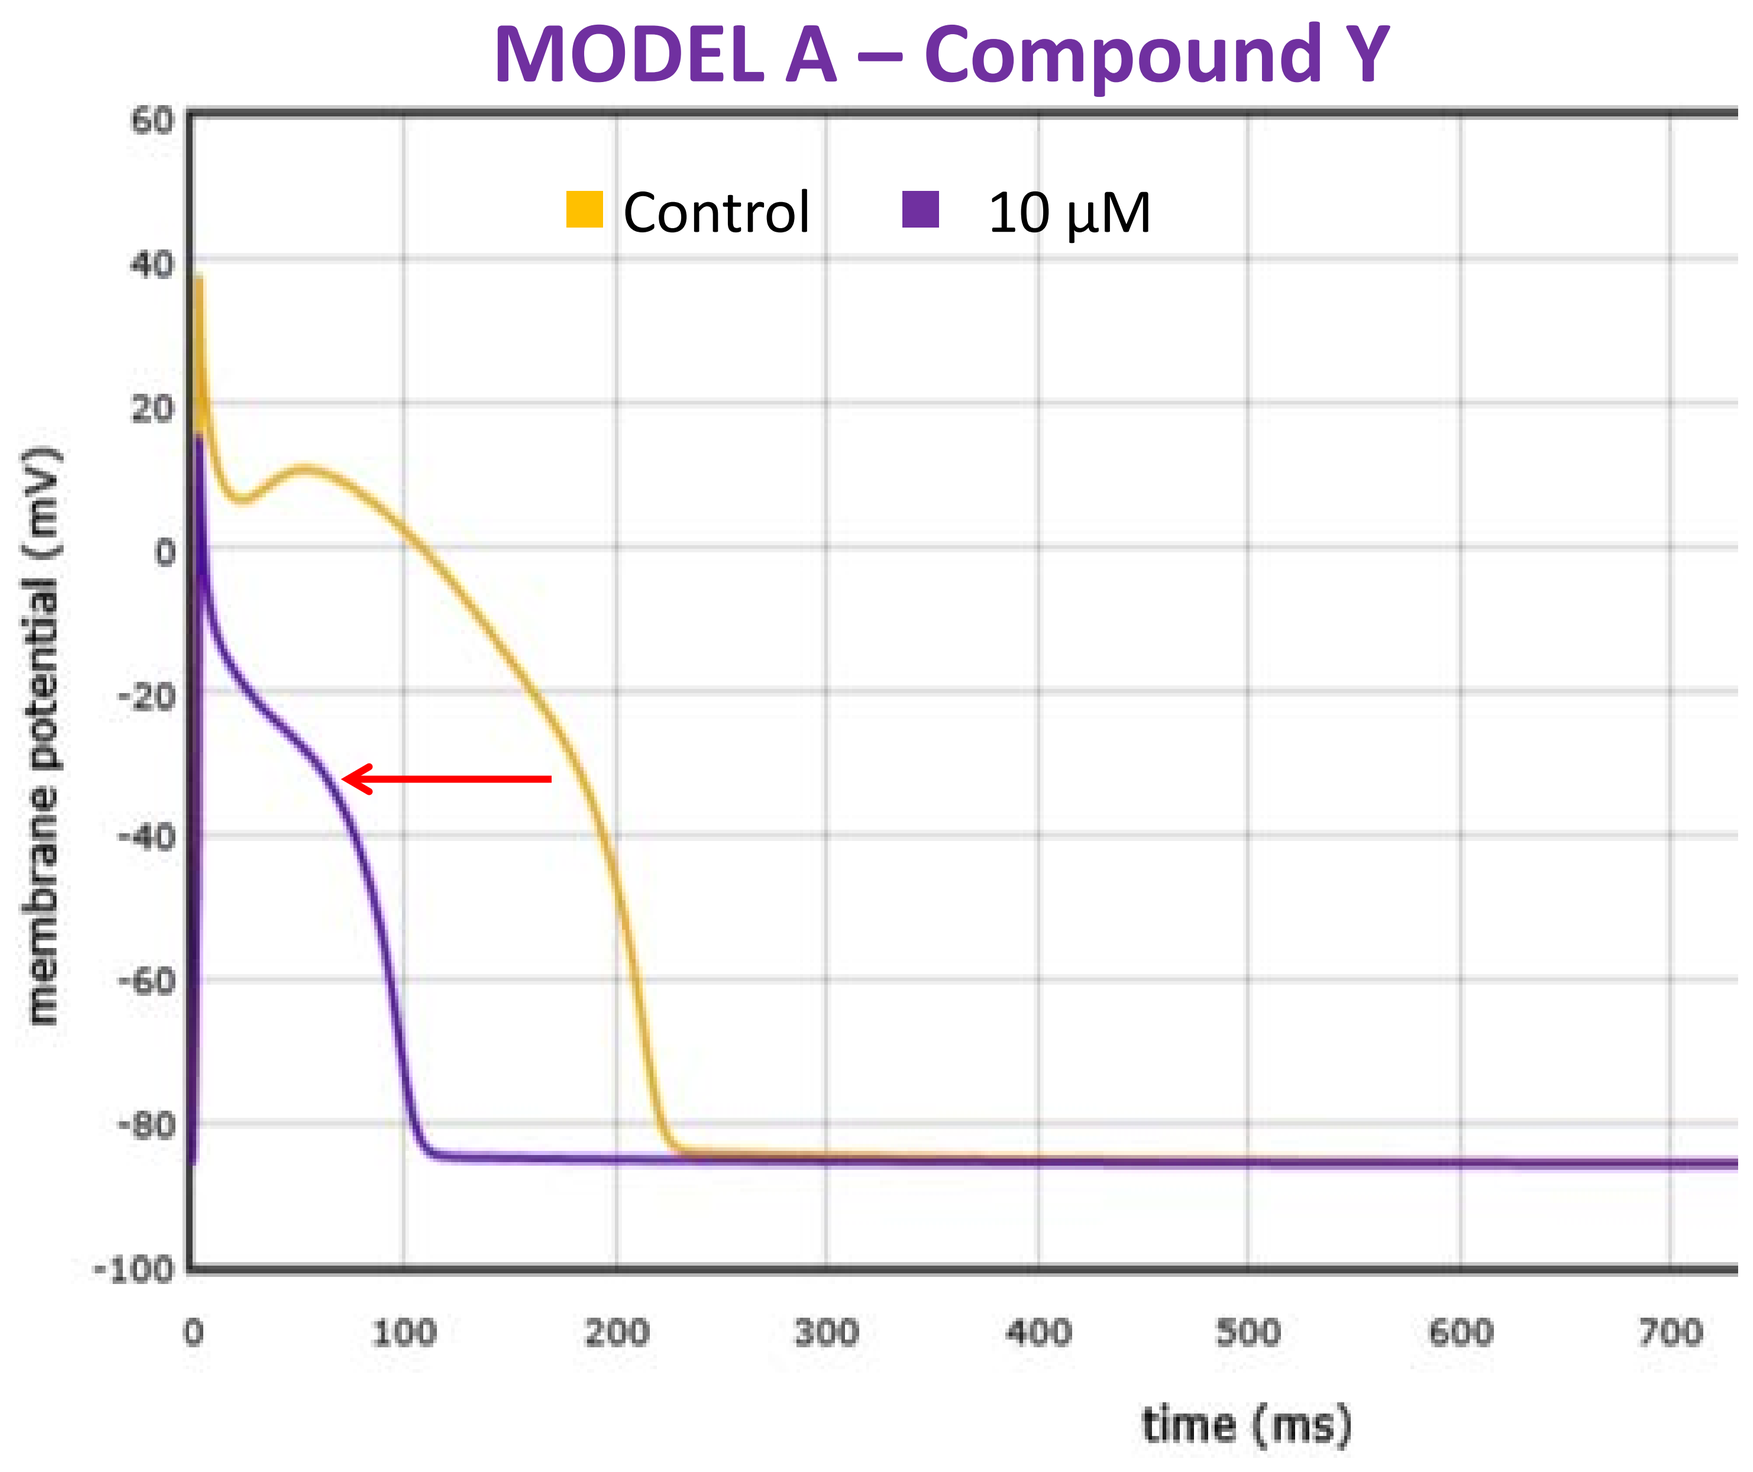
Control
10 µM
membrane potential (mV)
time (ms)

AP simulated in a dog ventricular myocyte :
62% decrease in APD_{90}— 67% decrease in APD_{50}

MODEL B – Compound Y

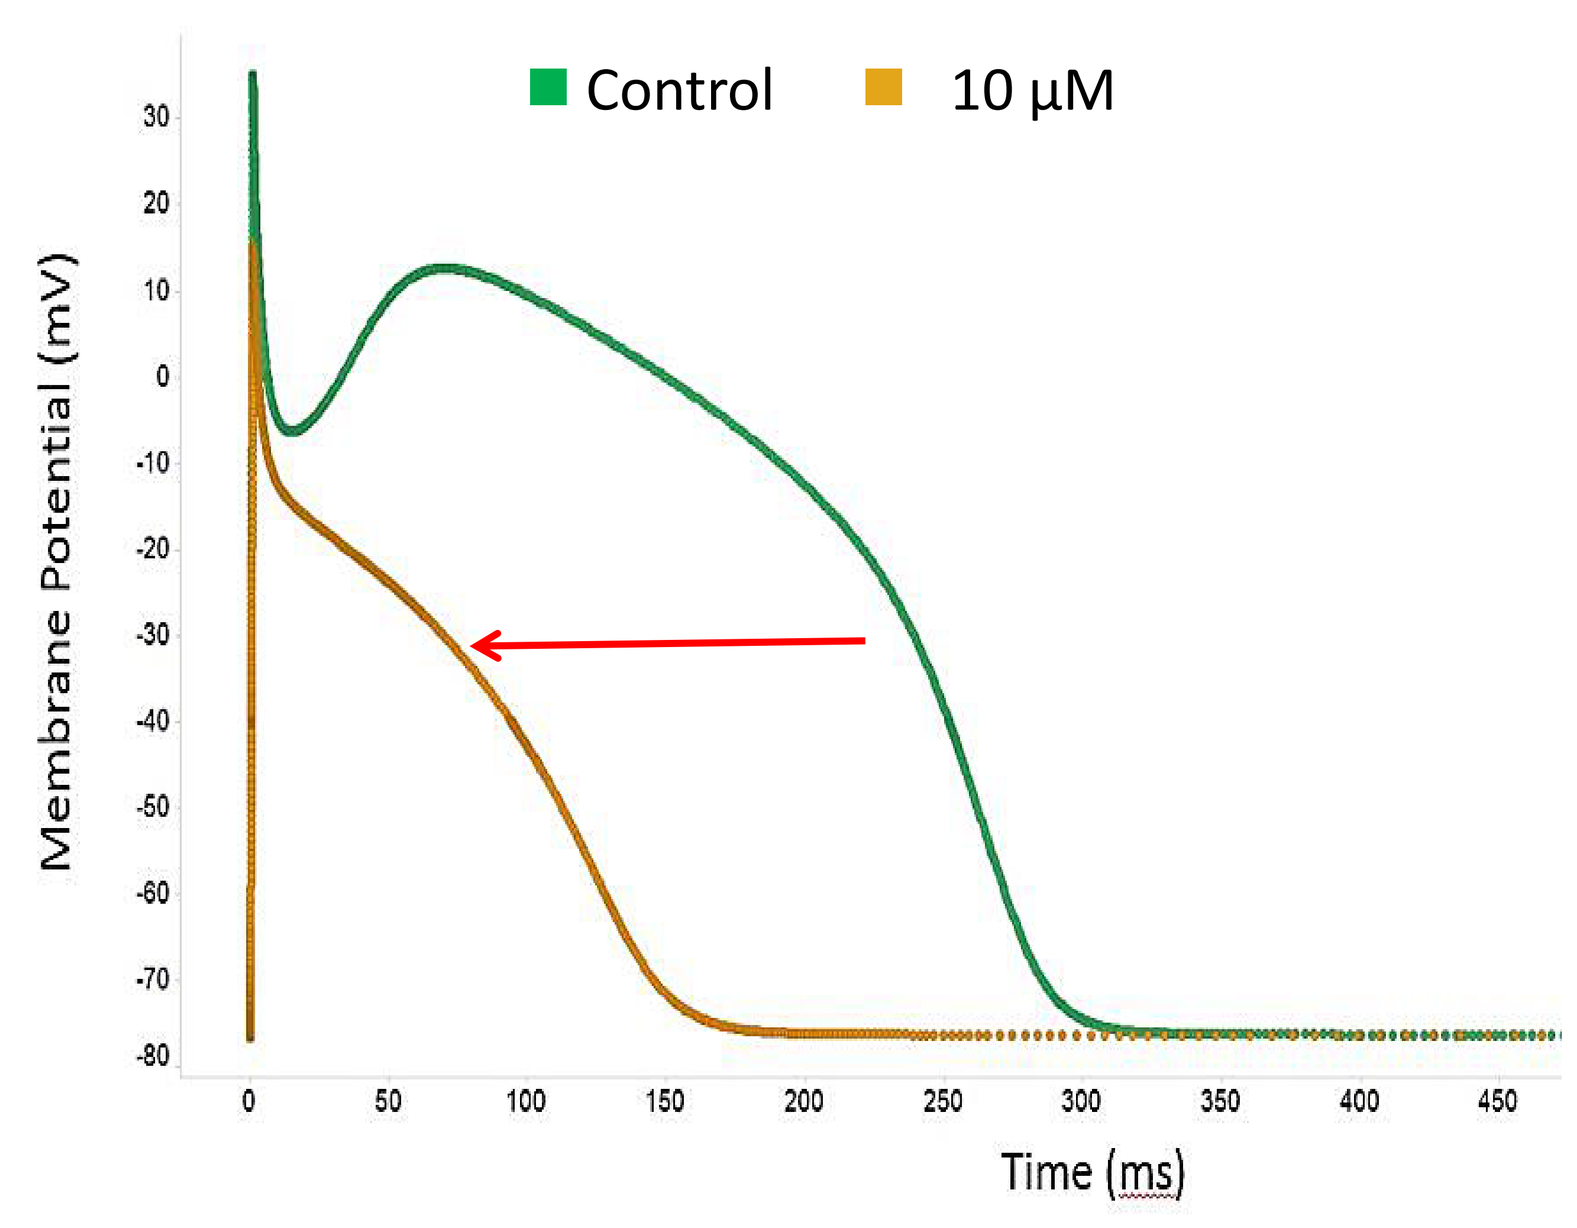
Control
10 µM
Membrane Potential (mV)
Time (ms)

AP simulated in a dog ventricular myocyte :
49% decrease in APD_{90}— 66 % decrease in APD_{50}

MODEL C – Compound Y

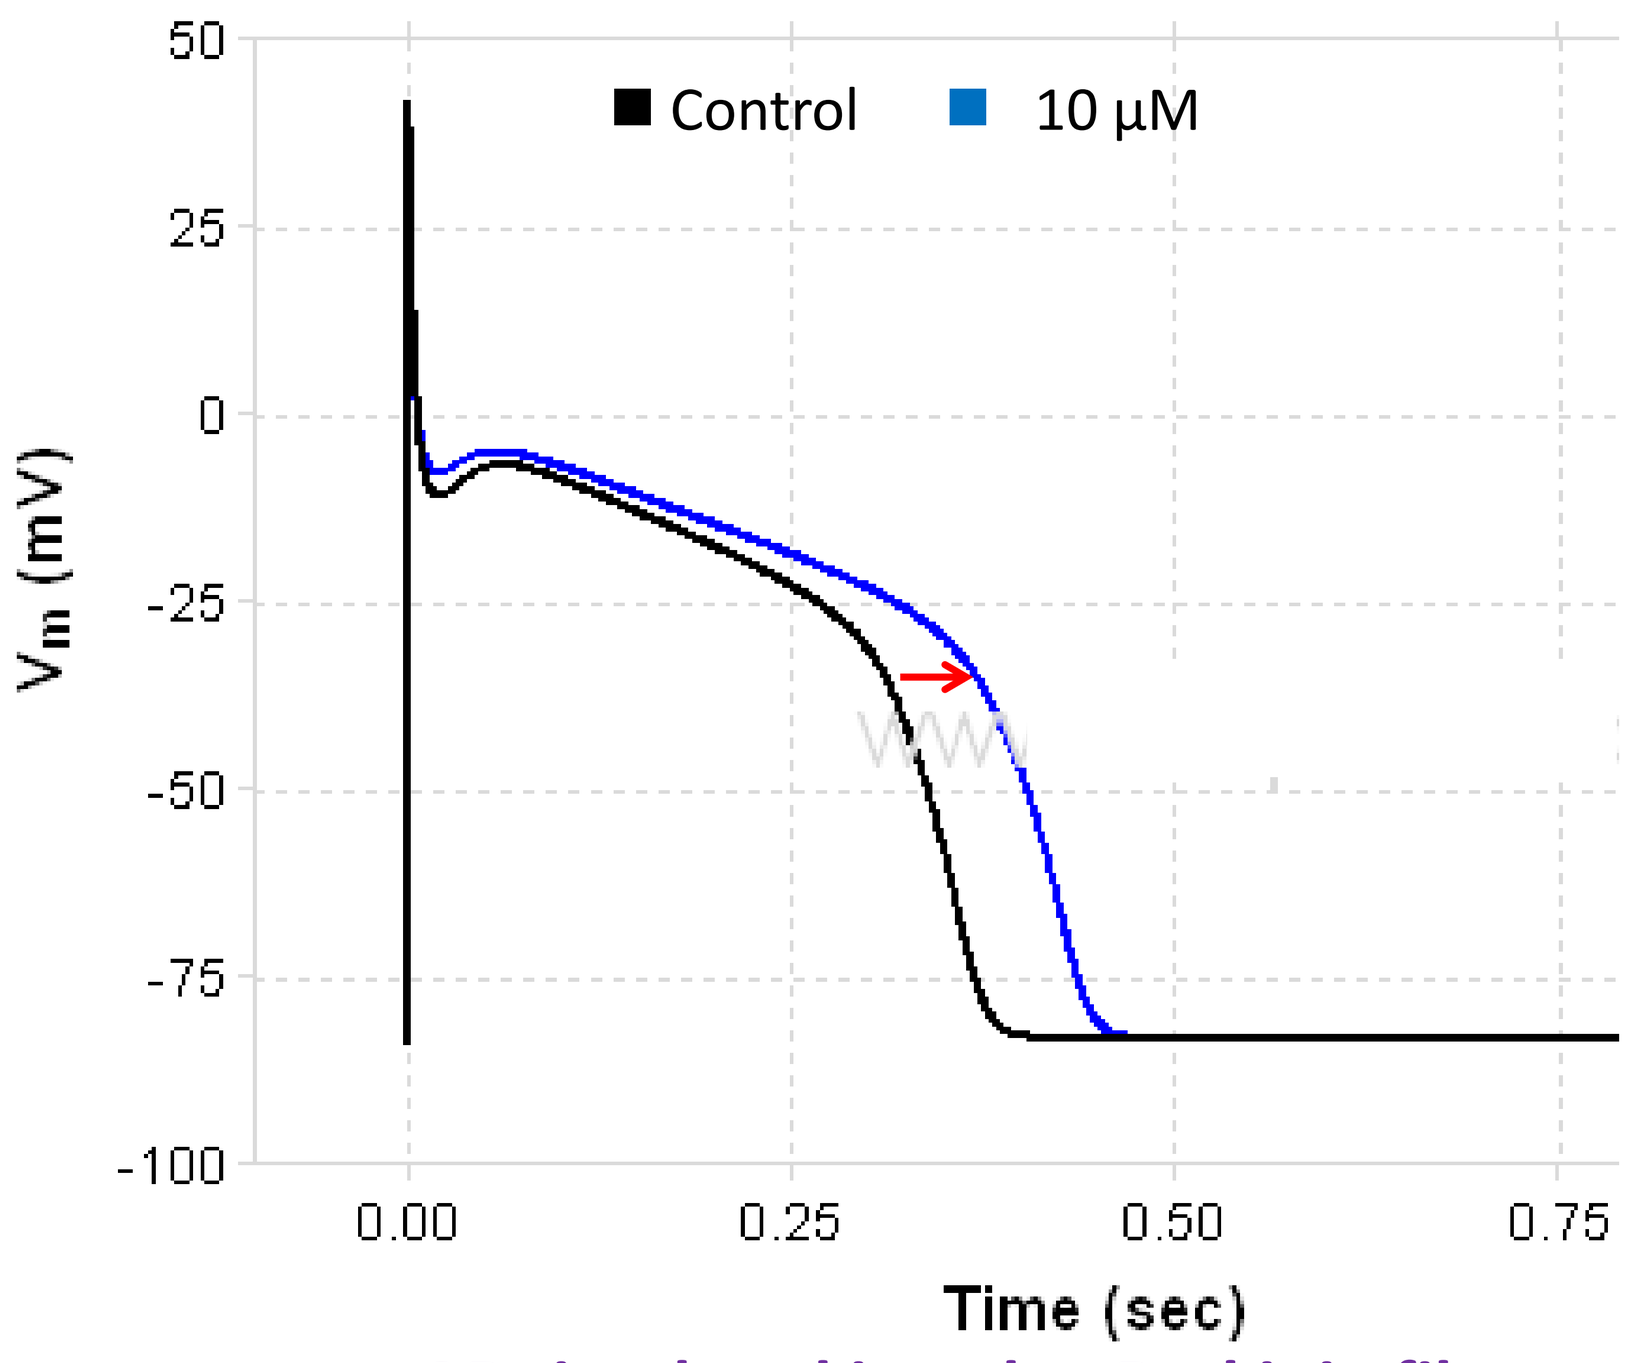
Control
10 µM
V_m (mV)
Time (sec)

AP simulated in a dog Purkinje fiber:
36% increase in APD_{90}— 277% increase in APD_{40}

REAL DATA – Compound Y

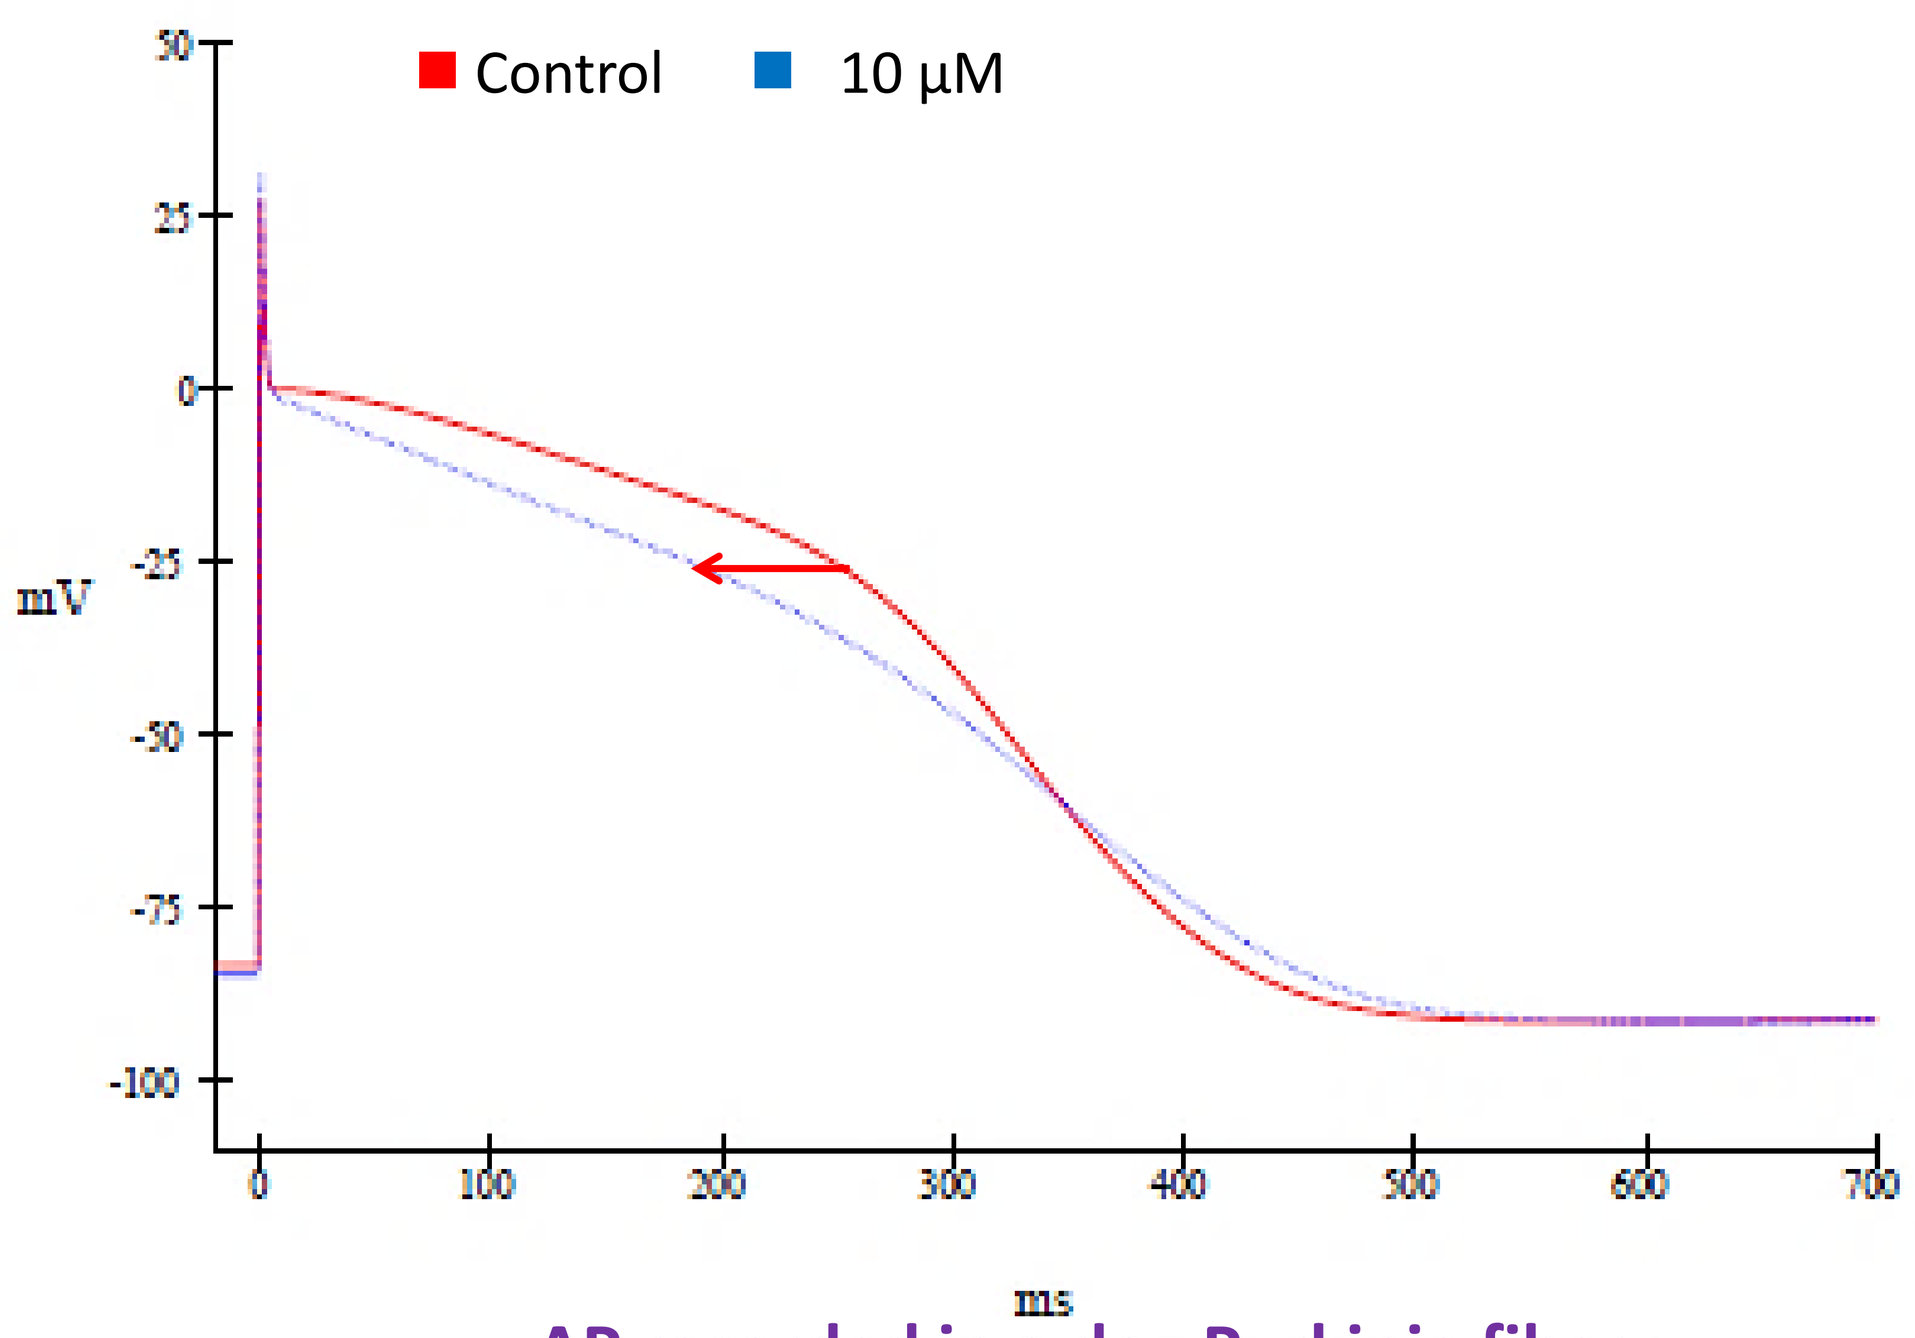
Control
10 µM
mV
ms

AP recorded in a dog Purkinje fiber :
3% increase in APD_{90}— 28 % decrease in APD_{50}

Conclusions

- Existing *in silico* models can considerably vary in terms of number of channels integrated, parameters generated, species and cell types selected,…As shown in the above example for Compound Y, results can also vary both qualitatively (nature of effects predicted) and quantitatively (magnitude or threshold of effects).
- In our comparison, only one tested model (B) successfully predicted all real world AP effects for the 3 compounds. Overall, *in silico* models appear more 'sensitive' than real world data, as predicted effects occur at lower concentrations than really measured
- Although *in silico* modeling of action potential effects appears as a promising tool to increase likelihood of success due to absence of pro-arrhythmia risk, a careful evaluation and validation of the model(s) selected is recommended before routine usage in cardiac safety screening strategy.

Inspired by patients.
Driven by science.